In Vitro Investigation of Masitinib as a Chemosensitizer in Canine Cancer

Douglas H. Thamm[1], Barbara Rose[1], Kelvin Kow[1], Martine Humbert[2], Alain Moussy[2], Olivier Hermine[3] and Patrice Dubreuil[4]

[1]The Animal Cancer Center, Colorado State University, Ft. Collins, CO, USA. [2]AB Science S.A., Paris, France. [3]CNRS UMR 8147, Assistance Publique Hôpitaux de Paris and Service d'Hématologie Adulte, Centre de référence sur la mastocytose, Hôpital Necker, Université Paris V, Paris, France. [4]Inserm, U891, Centre de Recherche en Cancérologie de Marseille, Molecular and Functional Hematopoiesis, Centre de référence des mastocytoses, Marseille, F-13009, France; Institut Paoli-Calmettes, Marseille, F-13009, France; Univ Méditerranée, F-13007, Marseille, France

1. ABSTRACT

We recently showed that masitinib, a tyrosine kinase inhibitor targeting c-Kit, is safe and effective for the treatment of grade II or III nonresectable or recurrent mast cell tumors in dogs. To investigate the possible use of masitinib in combination chemotherapy in dogs, we examined its ability to sensitize canine cancer cell lines to doxorubicin, vinblastine, and gemcitabine. Masitinib (0.001-10 uM) sensitized all canine cancer cell lines to doxorubicin (2- to 10-fold reduction in IC_{50}), strongly sensitized DH82 histiocytic sarcoma cells to vinblastine (>74-fold reduction), and strongly sensitized D17 and Abrams osteosarcoma and CMT12 and CMT27 mammary carcinoma cells to gemcitabine (>10-fold reduction). In summary, masitinib can chemosensitize canine tumor cell lines to chemotherapeutic agents. Thus, further investigation of the use of masitinib in combination chemotherapy in dogs is warranted.

Table 1

Name	Tumor Type	Originator
17CM98	Melanoma	G. Hogge, U . of Wisconsin
CML-6M	Melanoma	L. Wolfe, Auburn U
CML-10C2	Melanoma	L. Wolfe, Auburn U
Abrams	Osteosarcoma	E. G. MacEwen, U. of Wisconsin
D17	Osteosarcoma	ATCC
K9TCC	Bladder Carcinoma	D. Knapp, Purdue U.
Bliley	Bladder Carcinoma	S. Dow, Colorado State U.
DEN	Hemangiosarcoma	D. Thamm, Colorado State U.
Fitz	Hemangiosarcoma	D. Thamm, Colorado State U.
CMT12	Mammary Carcinoma	L. Wolfe, Auburn U
CMT27	Mammary Carcinoma	L. Wolfe, Auburn U
C2	Mast Cell Tumor	W. Gold, UCSF
1771	B Cell Lymphoma	K. A. Jeglum, U. of Pennsylvania
OSW	T Cell Lymphoma	W. Kisseberth, Ohio State U.
DH82	Histiocytic Sarcoma	ATCC

2. CONCLUSIONS

- Masitinib sensitizes canine tumor cells to the antiproliferative effects of multiple antineoplastic drugs at clinically achievable concentrations.
- The OSW-LSA T cell lymphoma cell line demonstrates unique single-agent sensitivity to masitinib, through an unknown mechanism. Studies are ongoing to determine whether other T cell malignancies are similarly sensitive.
- The encouraging data presented here strongly justify clinical evaluation of masitinib / chemotherapy combination treatment in dogs with spontaneous tumors.

In summary, we propose the following treatments with masitinib that may work the best for the different canine tumor types:

Canine tumor type	Suggested treatments
Mastocytoma T Cell Lymphoma	**Monotherapy:** **masitinib**
	Combinatory therapy: **masitinib + chemotherapy**
Osteosarcoma Mammary Carcinoma	masitinib + gemcitabine
Melanoma Hemangiosarcoma Bladder Carcinoma B Cell Lymphoma	masitinib + doxorubicin
Histiocytic Sarcoma	masitinib + vinblastine

3. SINGLE AGENT ASSAYS

Cell Lines and Conditions

The canine cell lines utilized in these experiments are listed in Table 1. With the exception of the C2, OSW and 1771 lines, cells were serially passaged by trypsinization on plastic in Minimal Essential Medium (MEM, BioWhittaker, Walkersville, MD) supplemented with 5% heat activated fetal bovine serum (HyClone, Logan, UT), 5% heat-inactivated newborn calf serum, 100 units/ml penicillin-streptomycin (Mediatech, Herndon, VA), 2 mM L-Glutamine (Mediatech), 1 mM sodium pyruvate (Mediatech) and 1X nonessential amino acid solution (Sigma, St. Louis, MO) under standard conditions (37°C, 5% CO_2 in a humidified atmosphere). The C2, OSW and 1771 lines grow in suspension and were passaged in the same medium by density gradient centrifugation.

Figure 1

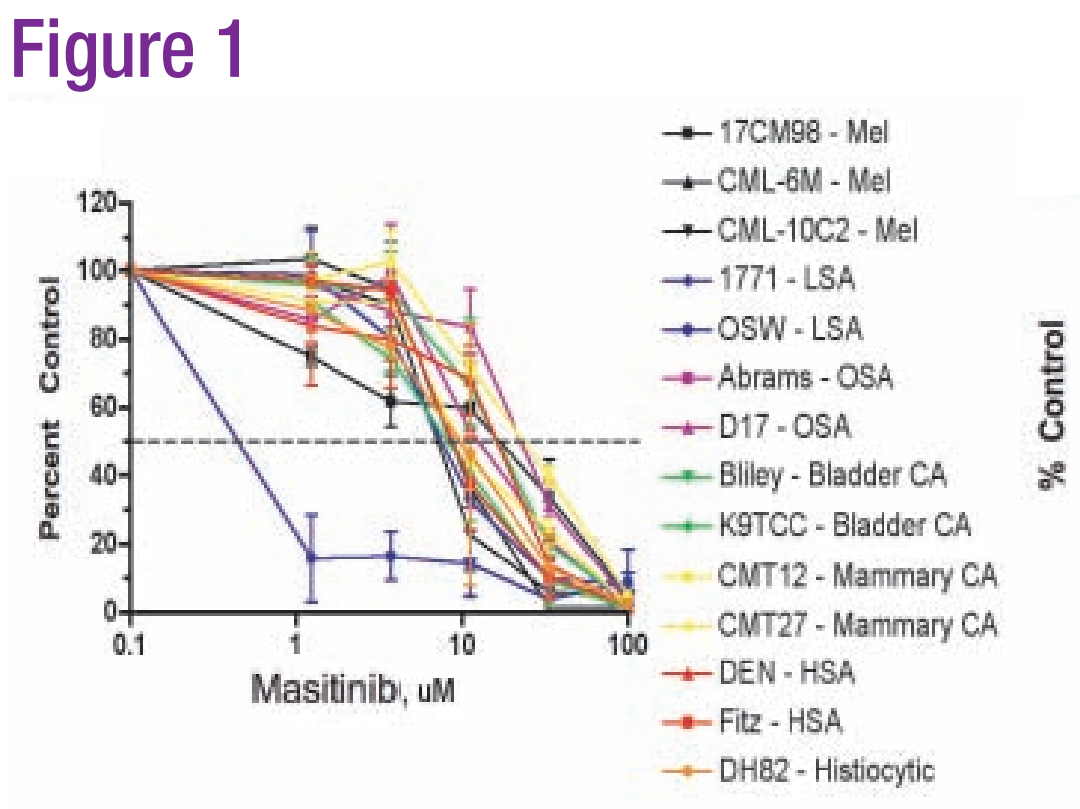
17CM98 - Mel
CML-6M - Mel
CML-10C2 - Mel
1771 - LSA
OSW - LSA
Abrams - OSA
D17 - OSA
Bliley - Bladder CA
K9TCC - Bladder CA
CMT12 - Mammary CA
CMT27 - Mammary CA
DEN - HSA
Fitz - HSA
DH82 - Histiocytic
Percent Control
Masitinib, uM

Figure 2

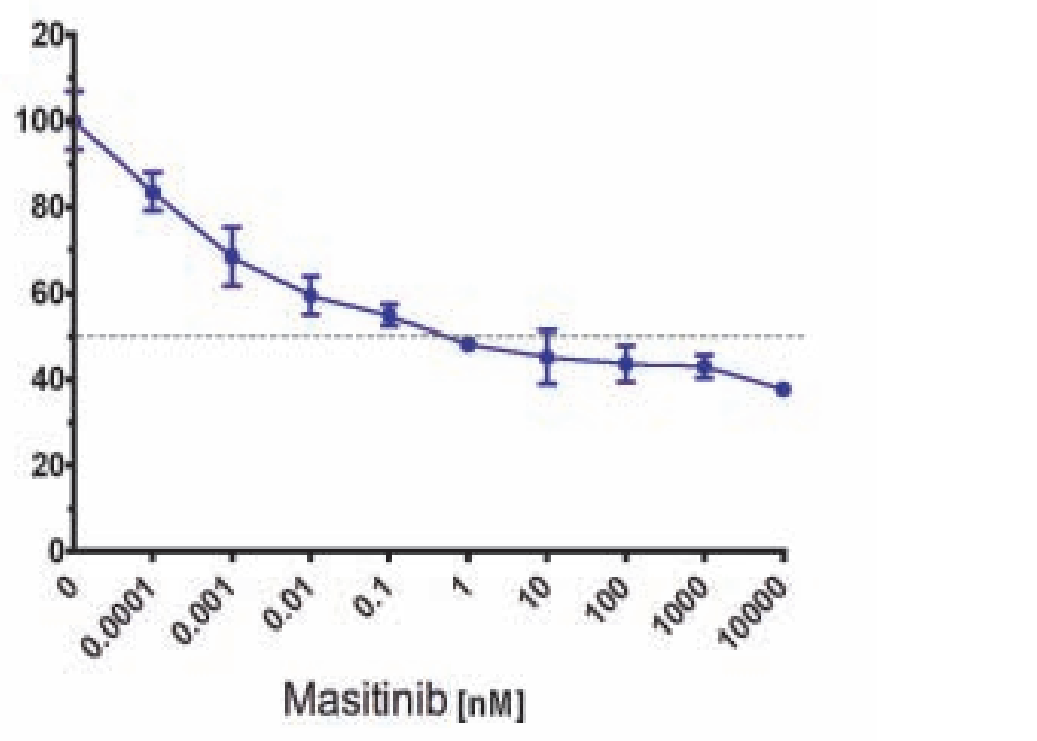
% Control
Masitinib [nM]

Figure 1, 2 Single-agent antiproliferative activity of masitinib against canine tumor cells.

1. Cells were plated in 96-well plates and allowed to adhere overnight.The following day, the plates were washed and 10% serum containing medium increasing concentrations of masitinib were added. 72 hours later, relative viable cell number was determined using a bioreductive fluorometric assay (Alamar Blue™). Cell number was standardized to cells incubated without masitinib. **1.** *Masitinib dose-dependently inhibited canine tumor cell growth, with 50% inhibitory concentrations of approximately 10 to 30 uM.* **2.** Extreme masitinib sensitivity was documented in OSW T cell lymphoma cells, with an IC_{50} of approximately 1 nM.

4. CHEMOSENSITIVITY ASSAYS

To assess the effect of varying concentrations of masitinib on sensitivity to common antineoplastic agents, anchorage-dependent cells were plated at varying cell numbers (based on prior optimization) in 96-well plates in 10% serum-containing complete MEM (C/10) and allowed to adhere overnight. The following day, the plates were washed and the media replaced with C/10 containing varying concentrations of chemotherapeutic agent and 2 different concentrations of masitinib. Concentrations of chemotherapeutic agent and masitinib used were established based on preliminary experiments. Suspension (non-adherent) cells were treated similarly, with the exception of the overnight initial plating. After a 72-hour incubation under standard conditions, relative viable cell number was assessed utilizing a fluorescent bioreductive assay (Alamar Blue, Promega, Madison, WI) according to manufacturer specifications. Cell growth was expressed as a percentage of control cells incubated in C/10 +/- masitinib without chemotherapy. Each condition was performed in triplicate. 50% inhibitory concentrations (IC_{50}s) for chemotherapy with and without masitinib were calculated using nonlinear regression: data were fitted to a sigmoidal dose-response curve. Each experiment was repeated at least 3 times.

4.1 DOXORUBICIN SENSITIVITY

Figure 3

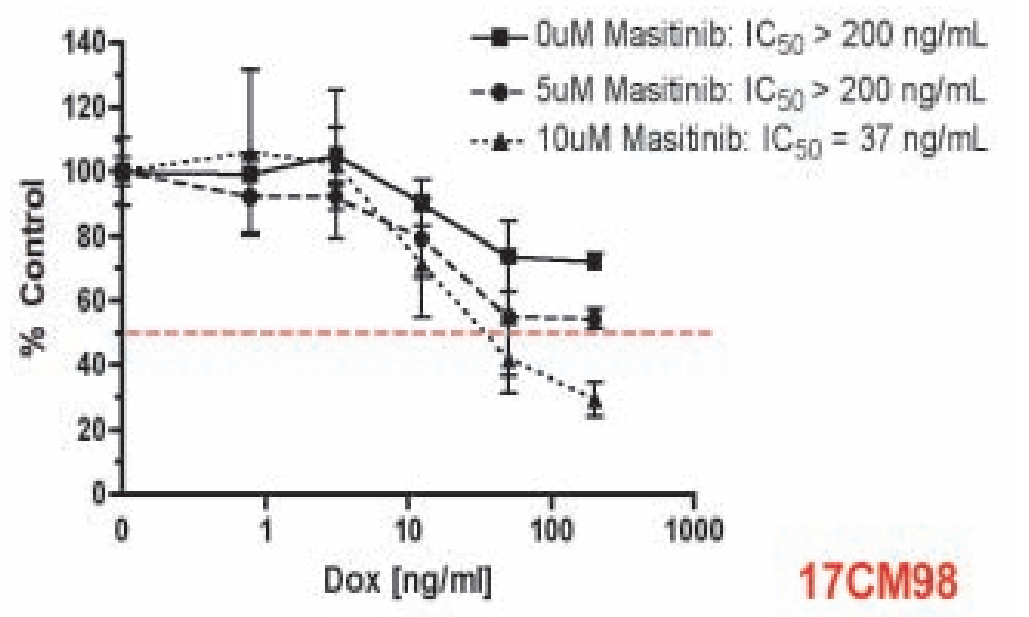
0uM Masitinib: IC50 > 200 ng/mL
5uM Masitinib: IC50 > 200 ng/mL
10uM Masitinib: IC50 = 37 ng/mL
% Control
Dox [ng/ml]
17CM98

Figure 4

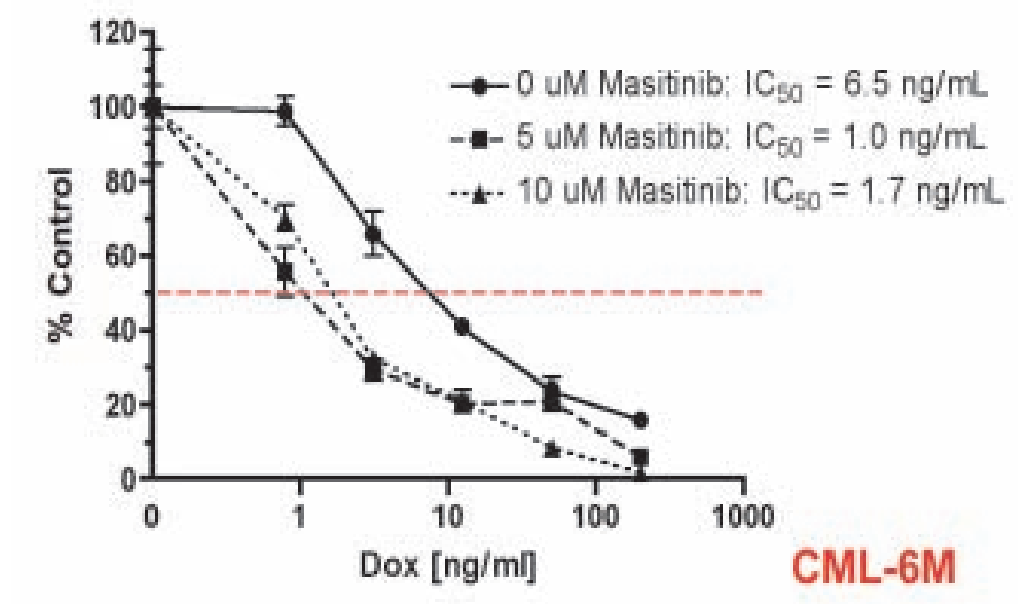
0 uM Masitinib: IC50 = 6.5 ng/mL
5 uM Masitinib: IC50 = 1.0 ng/mL
10 uM Masitinib: IC50 = 1.7 ng/mL
% Control
Dox [ng/ml]
CML-6M

Figure 5

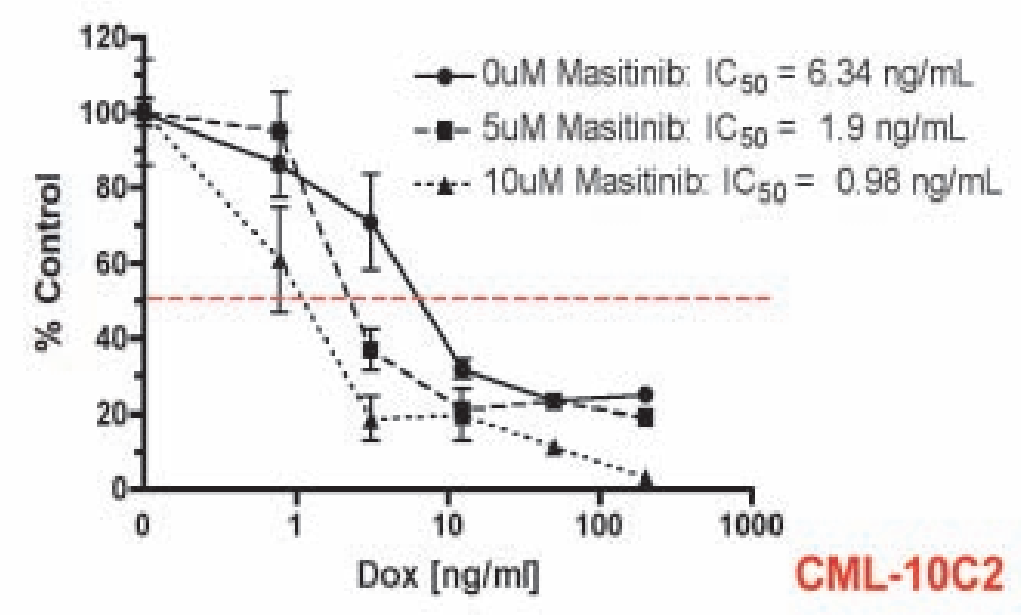
0uM Masitinib: IC50 = 6.34 ng/mL
5uM Masitinib: IC50 = 1.9 ng/mL
10uM Masitinib: IC50 = 0.98 ng/mL
% Control
Dox [ng/ml]
CML-10C2

Figure 3, 4, 5 Masitinib sensitizes canine melanoma cells to doxorubicin. When co-incubated for 72 hours, masitinib reduced the doxorubicin IC_{50} by means of 4.9 and 5.1 fold for 5 uM and 10 uM concentrations, respectively.

Figure 6

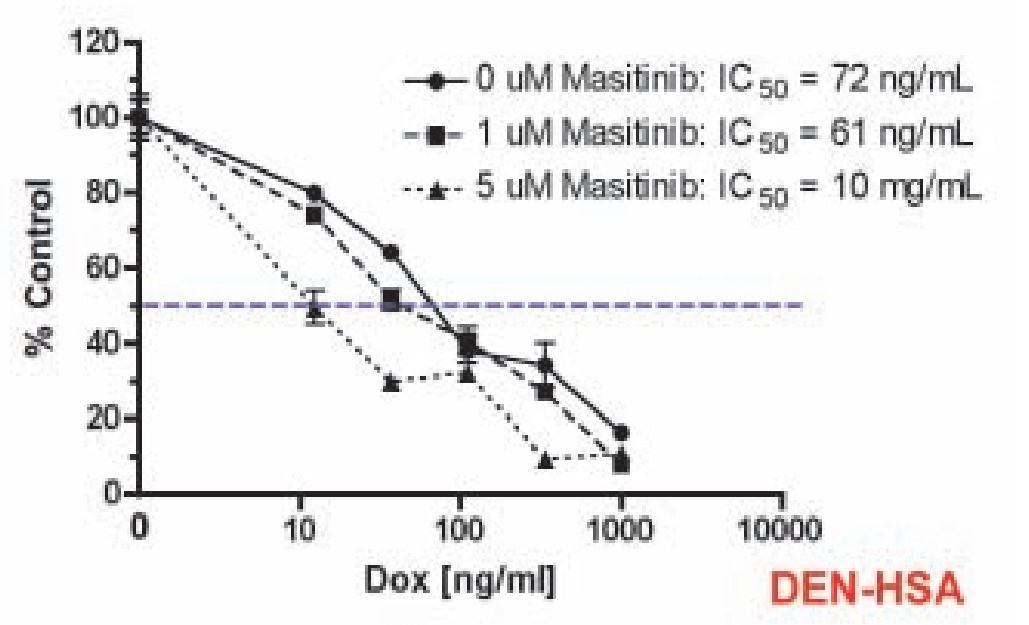
0 uM Masitinib: IC 50 = 72 ng/mL
1 uM Masitinib: IC 50 = 61 ng/mL
5 uM Masitinib: IC 50 = 10 mg/mL
% Control
Dox [ng/ml]
DEN-HSA

Figure 7

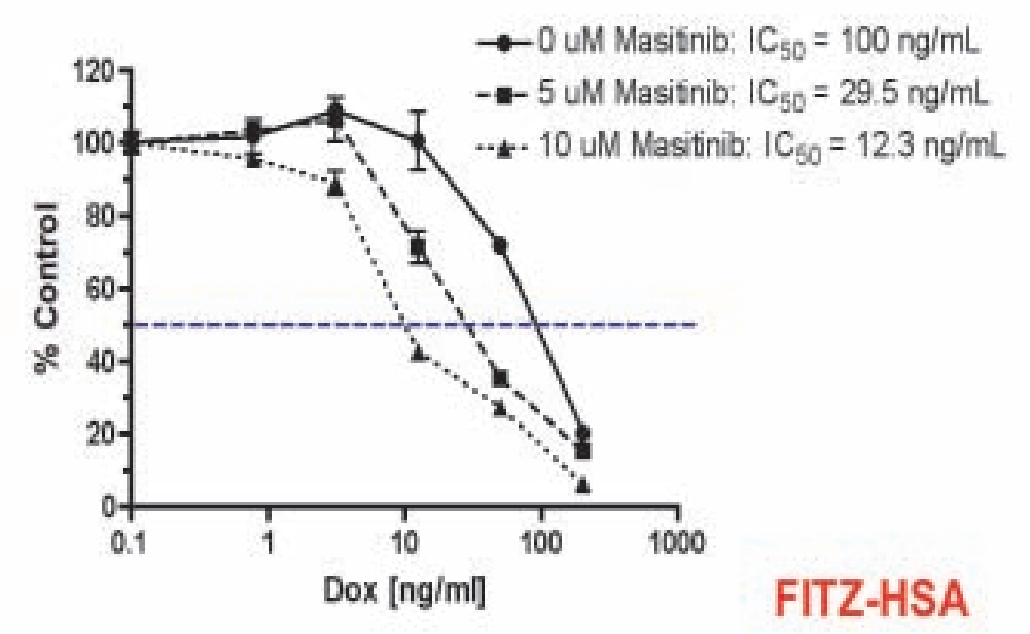
0 uM Masitinib: IC50 = 100 ng/mL
5 uM Masitinib: IC50 = 29.5 ng/mL
10 uM Masitinib: IC50 = 12.3 ng/mL
% Control
Dox [ng/ml]
FITZ-HSA

Figure 6,7 Masitinib sensitizes canine hemangiosarcoma to doxorubicin. When co-incubated for 72 hours, masitinib reduced the doxorubicin IC_{50} by means of 2.3 and 7.7 fold for low and high concentrations, respectively.

4.2 VINBLASTINE SENSITIVITY

Figure 8

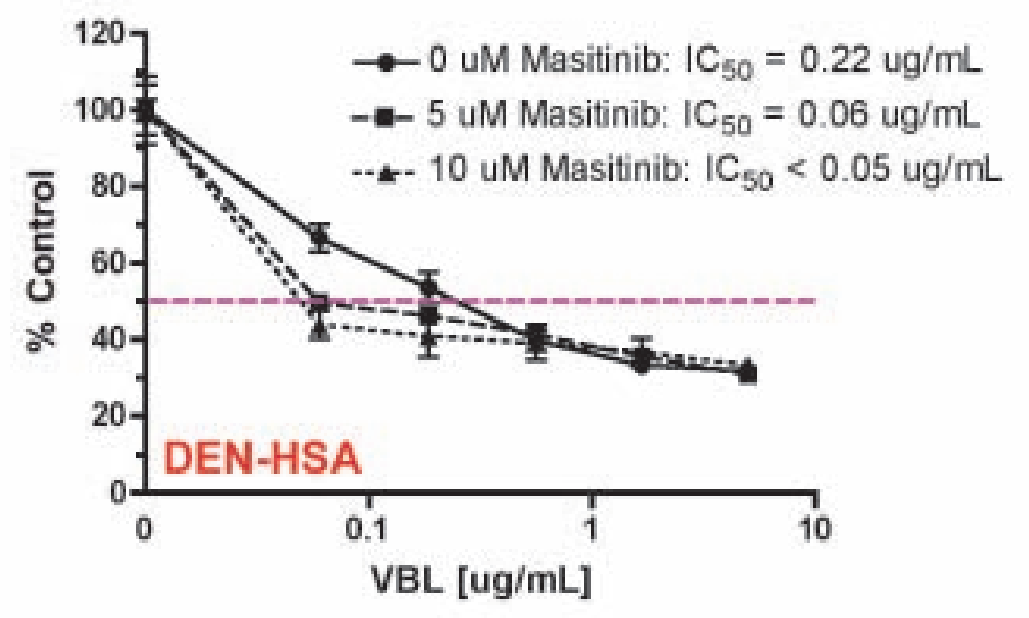
0 uM Masitinib: IC50 = 0.22 ug/mL
5 uM Masitinib: IC50 = 0.06 ug/mL
10 uM Masitinib: IC50 < 0.05 ug/mL
% Control
DEN-HSA
VBL [ug/mL]

Figure 9

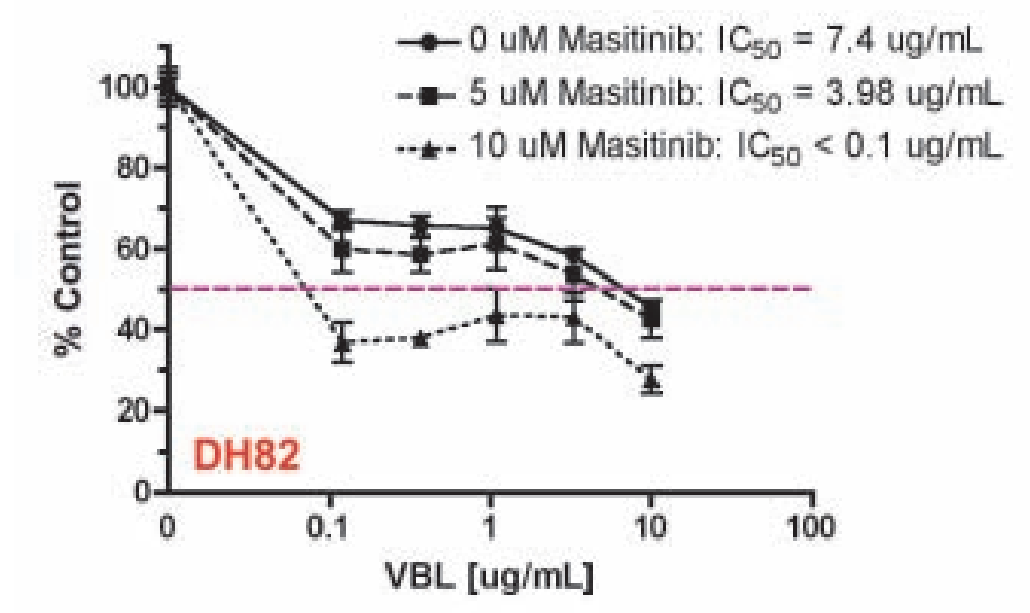
0 uM Masitinib: IC50 = 7.4 ug/mL
5 uM Masitinib: IC50 = 3.98 ug/mL
10 uM Masitinib: IC50 < 0.1 ug/mL
% Control
DH82
VBL [ug/mL]

Figure 8, 9 Masitinib sensitizes canine tumor cells to vinblastine. When co-incubated for 72 hours, masitinib reduced the vinblastine IC_{50} 4 to 70-fold for a concentration of 10 uM masitinib.

4.3 GEMCITABINE SENSITIVITY

Figure 10

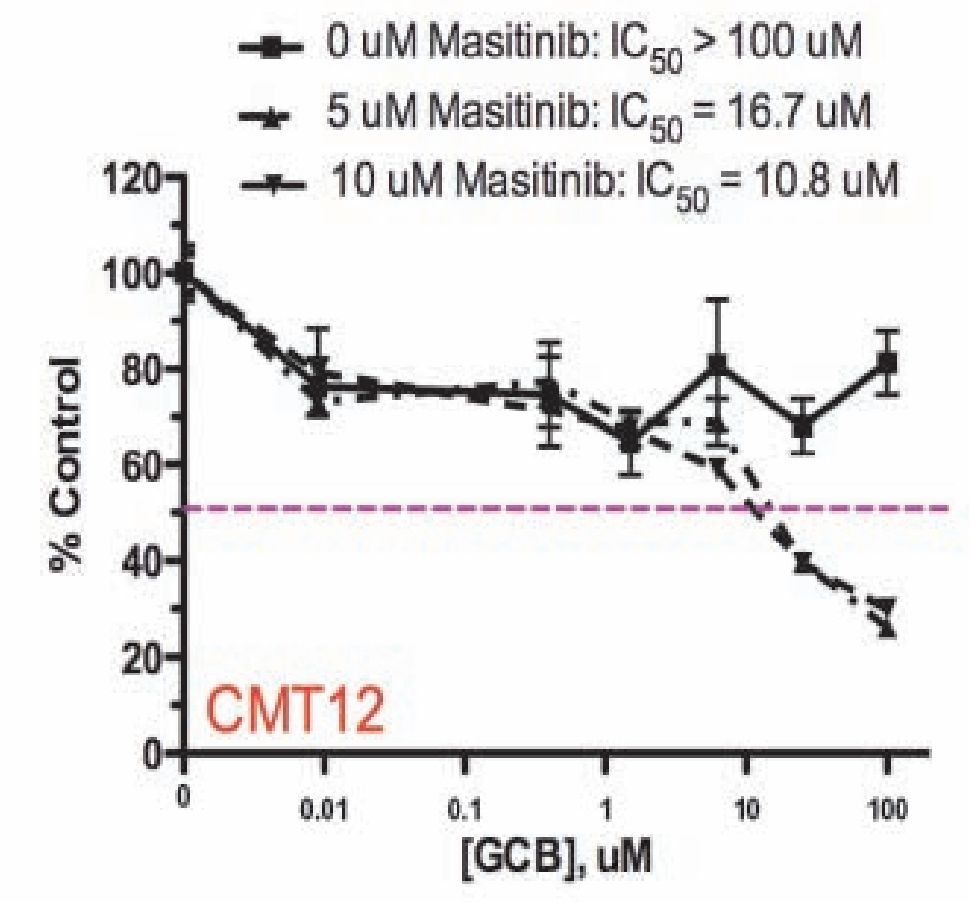
0 uM Masitinib: IC50 > 100 uM
5 uM Masitinib: IC50 = 16.7 uM
10 uM Masitinib: IC50 = 10.8 uM
% Control
CMT12
[GCB], uM

Figure 11

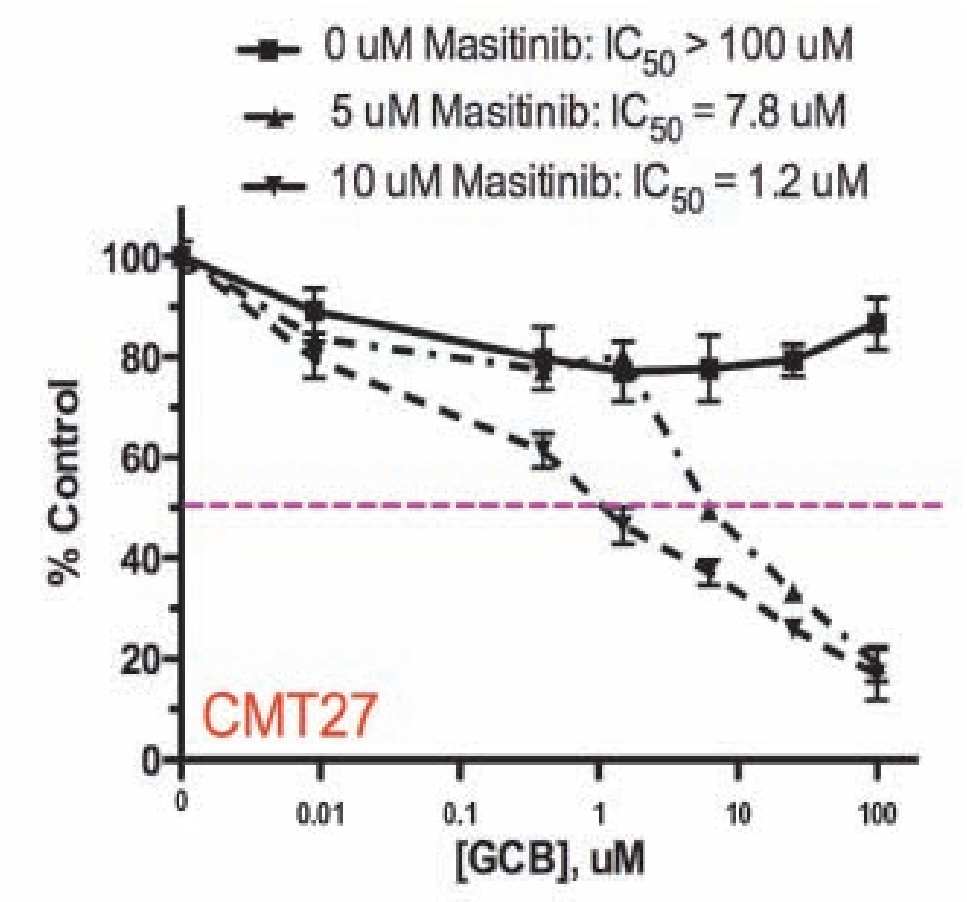
0 uM Masitinib: IC50 > 100 uM
5 uM Masitinib: IC50 = 7.8 uM
10 uM Masitinib: IC50 = 1.2 uM
% Control
CMT27
[GCB], uM

Figure 12

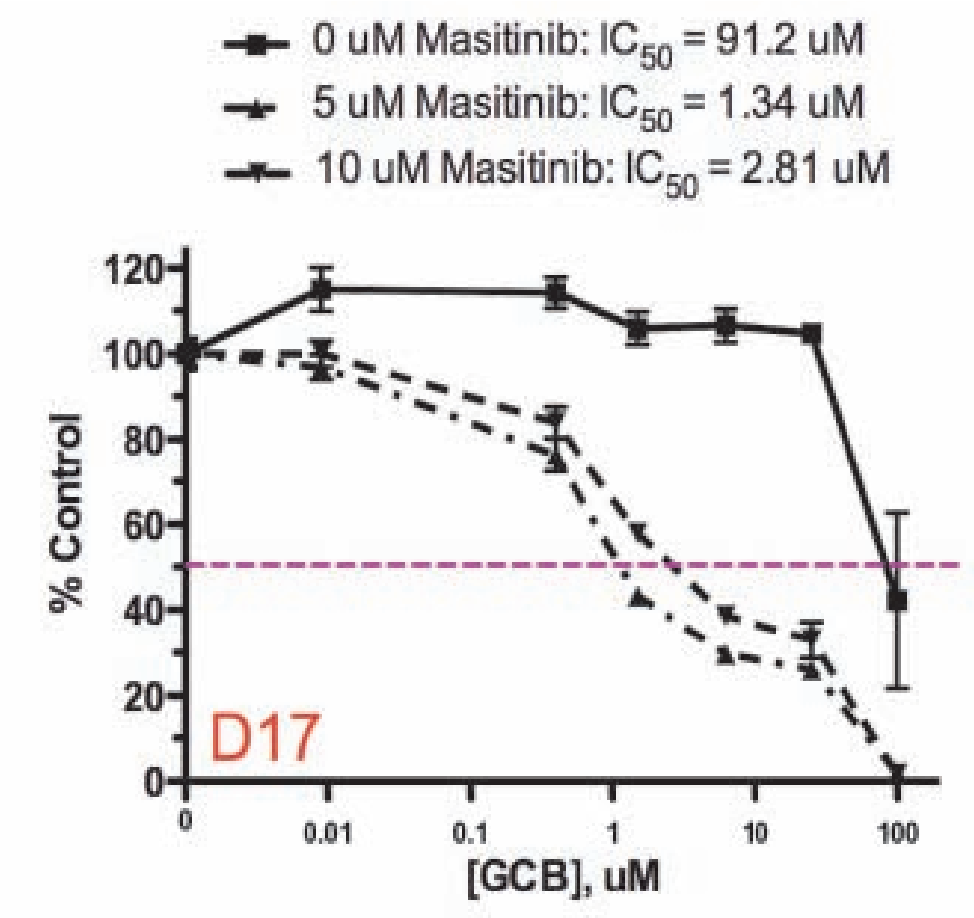
0 uM Masitinib: IC50 = 91.2 uM
5 uM Masitinib: IC50 = 1.34 uM
10 uM Masitinib: IC50 = 2.81 uM
% Control
D17
[GCB], uM

Figure 13

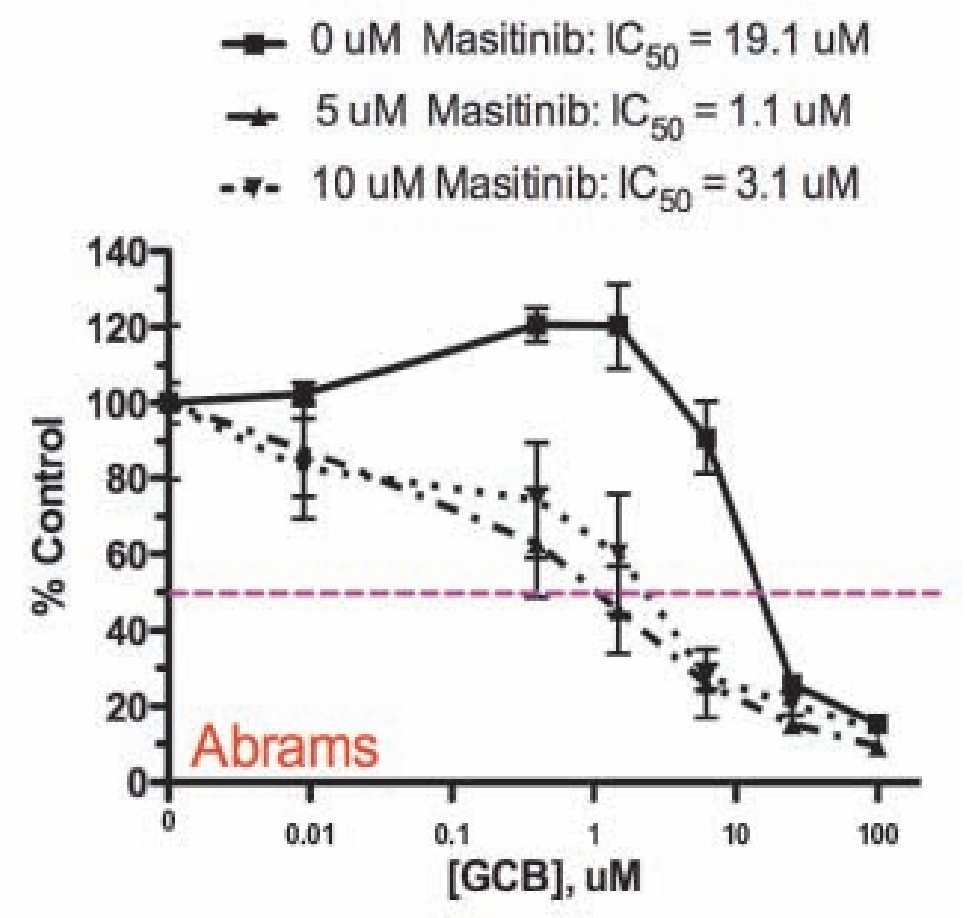
0 uM Masitinib: IC50 = 19.1 uM
5 uM Masitinib: IC50 = 1.1 uM
10 uM Masitinib: IC50 = 3.1 uM
% Control
Abrams
[GCB], uM

Figure 10, 11, 12, 13 Masitinib sensitizes canine tumor cells to gemcitabine. When co-incubated for 72 hours, masitinib reduced the gemcitabine IC_{50} 18 to > 75-fold for a concentration of 10 uM masitinib.

5. CELL LINES SENSITIVITY TO TREATMENT

Canine tumor type	IC_{50} Masitinib	IC_{50} Chemo	IC_{50} Chemo (+Masitinib)	SI (max)	Suggested treatments
Mastocytoma C2	0.03				**Monotherapy:** masitinib
T cell Lymphoma Oswald	0.005				**Monotherapy:** masitinib
Osteosarcoma Abrams D17	>10 >10	18 91.2	1.1 1.34	17 68	**Combinatory therapy:** masitinib + gemcitabine
Mammary Carcinoma CMT12 CMT27	8 8	>100 >100	10.8 1.3	>9 >78	**Combinatory therapy:** masitinib + gemcitabine
Melanoma 17CM98 CML-6M CML-10C2	16.8 6.5 7.5	>200 6.6 6.3	35.5 1 1	>6 6 6	**Combinatory therapy:** masitinib + doxorubicin
HEMANGIOSARCOMA DEN FITZ	>5 10	72.4 97.7	10 10	7 10	**Combinatory therapy:** masitinib + doxorubicin
Bladder Carcinoma K9TCC Bliley	8 >10	45.7 26.3	4.4 9.8	10 3	**Combinatory therapy:** masitinib + doxorubicin
B cell Lymphoma 1771	6	20.4	10.2	2	**Combinatory therapy:** masitinib + doxorubicin
Histiocytic Sarcoma DH82	>10	7.41	<0.1	>74	**Combinatory therapy:** masitinib + vinblastine

* SI=sensitization index represents the ratio between the IC_{50} of chemotherapy used alone and the IC_{50} of chemotherapy used in combination with masitinib

AB SCIENCE

Animal Cancer Center
Colorado State University Veterinary Teaching Hospital
Colorado State University